CAR T-cells targeting the kappa myeloma antigen for the treatment of multiple myeloma

Kavitha Gowrishankar[1], Rosanne Dunn[2], David Gottlieb[1], Kenneth Micklethwaite[1]

[1]University of Sydney, Sydney NSW, Australia; [2]HaemaLogiX Pty Ltd, Sydney, Australia

kavitha.gowrishankar@sydney.edu.au

#3572

DO NOT POST

INTRODUCTION

Multiple Myeloma (MM) is a malignancy of clonal plasma cells in the bone marrow. Despite recent advances in the treatment and management of MM, the majority of patients will ultimately relapse and die from their disease within 3-5 years.

Chimeric antigen receptor (CAR) T-cells for haematological malignancies are revolutionising immunotherapy for cancer, after dramatic response rates were obtained with CAR19 against B-cell leukaemia and lymphomas.

CAR T-cells for MM – There have been recent reports of phenomenal success with CARs targeting BCMA; other preclinical studies reported targeting CS1/SLAMF7, CD44v6, CD138, CD38, CD70.

KappaMab (MDX-1097) is a chimeric monoclonal antibody currently being assessed, in a Phase IIb clinical trial for the treatment of kappa light-chain restricted (κ-type) MM. (Trial ID - ACTRN12616001164482). MDX-1097 binds to a conformational epitope on **kappa myeloma antigen (KMA)**, a tumour-specific membrane associated protein expressed on malignant plasma cells in patients with κ-type MM (and light chains not associated with Immunoglobulin heavy chains).

METHODOLOGY

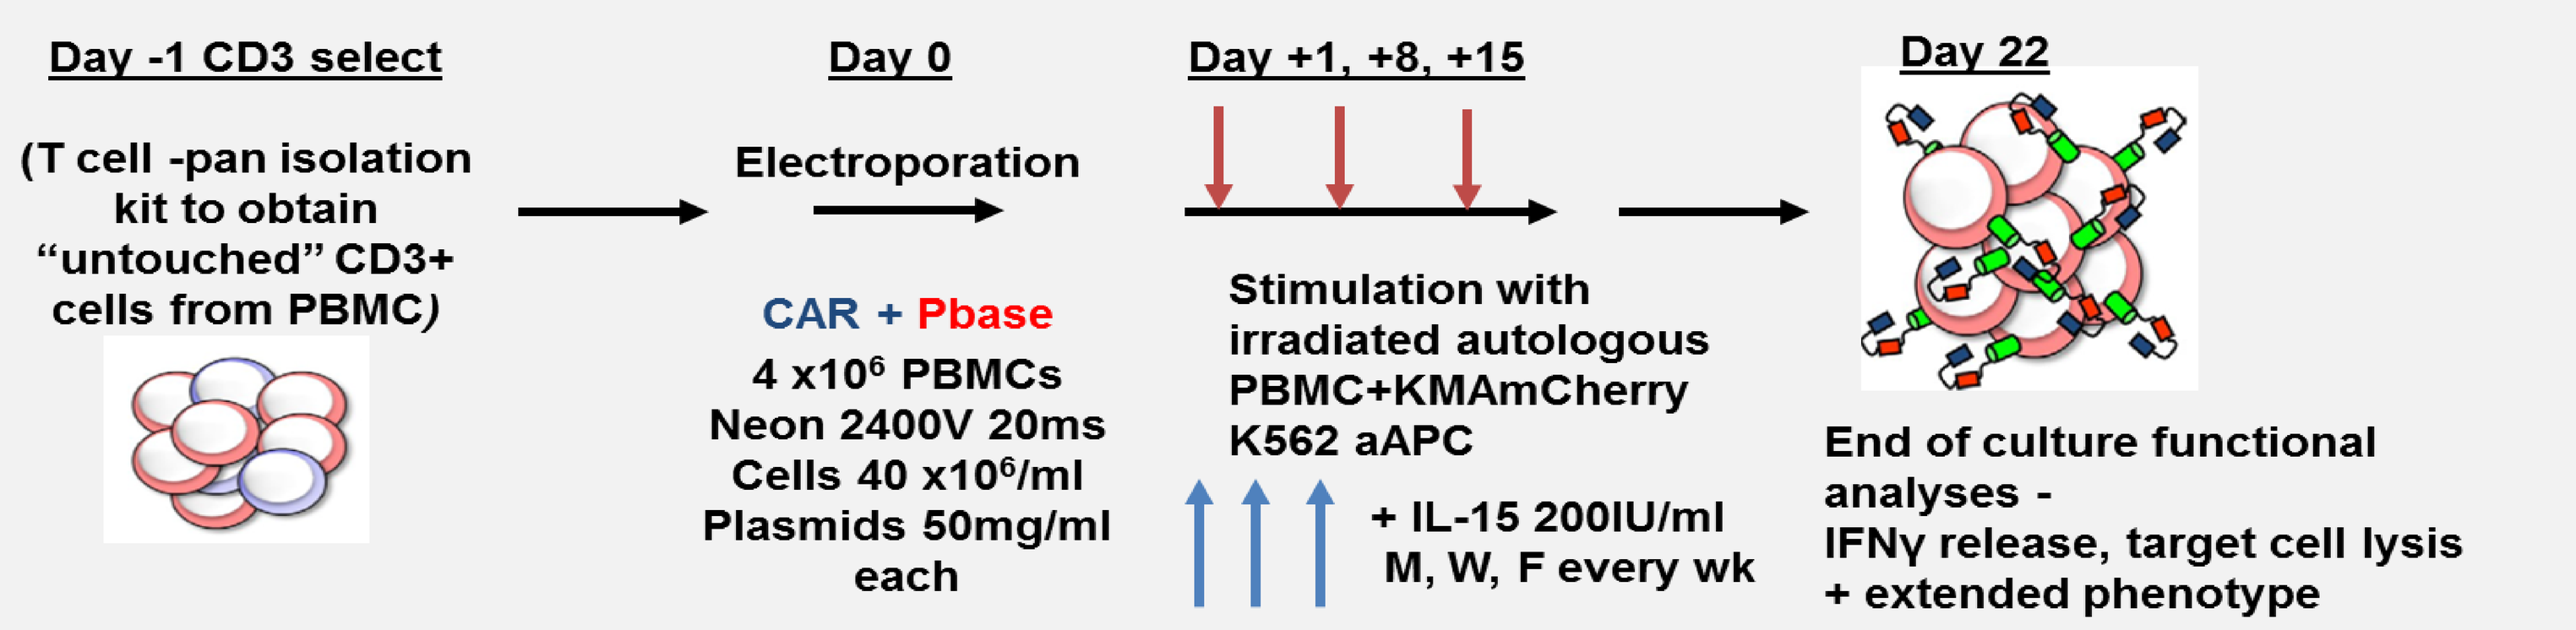
Day -1 CD3 select
(T cell -pan isolation kit to obtain "untouched" CD3+ cells from PBMC)
Day 0
Electroporation
CAR + Pbase
4 x10⁶ PBMCs
Neon 2400V 20ms
Cells 40 x10⁶/ml
Plasmids 50mg/ml each
Day +1, +8, +15
Stimulation with irradiated autologous PBMC+KMAmCherry K562 aAPC
+ IL-15 200IU/ml
M, W, F every wk
Day 22
End of culture functional analyses -
IFNγ release, target cell lysis
+ extended phenotype

CD3+ cells were isolated by MACS separation and co-electroporated with CAR piggyBac transposon DNA and piggyBac transposase DNA using the Neon system (Invitrogen). Transfected CAR T-cells were preferentially expanded by stimulation at days 1, 8 and 15 post-transfection with irradiated PBMC, irradiated KMA expressing K562 cell line (aAPC) and IL-15. Phenotyping and functional characterisation of CAR T-cells were carried out at end of culture.

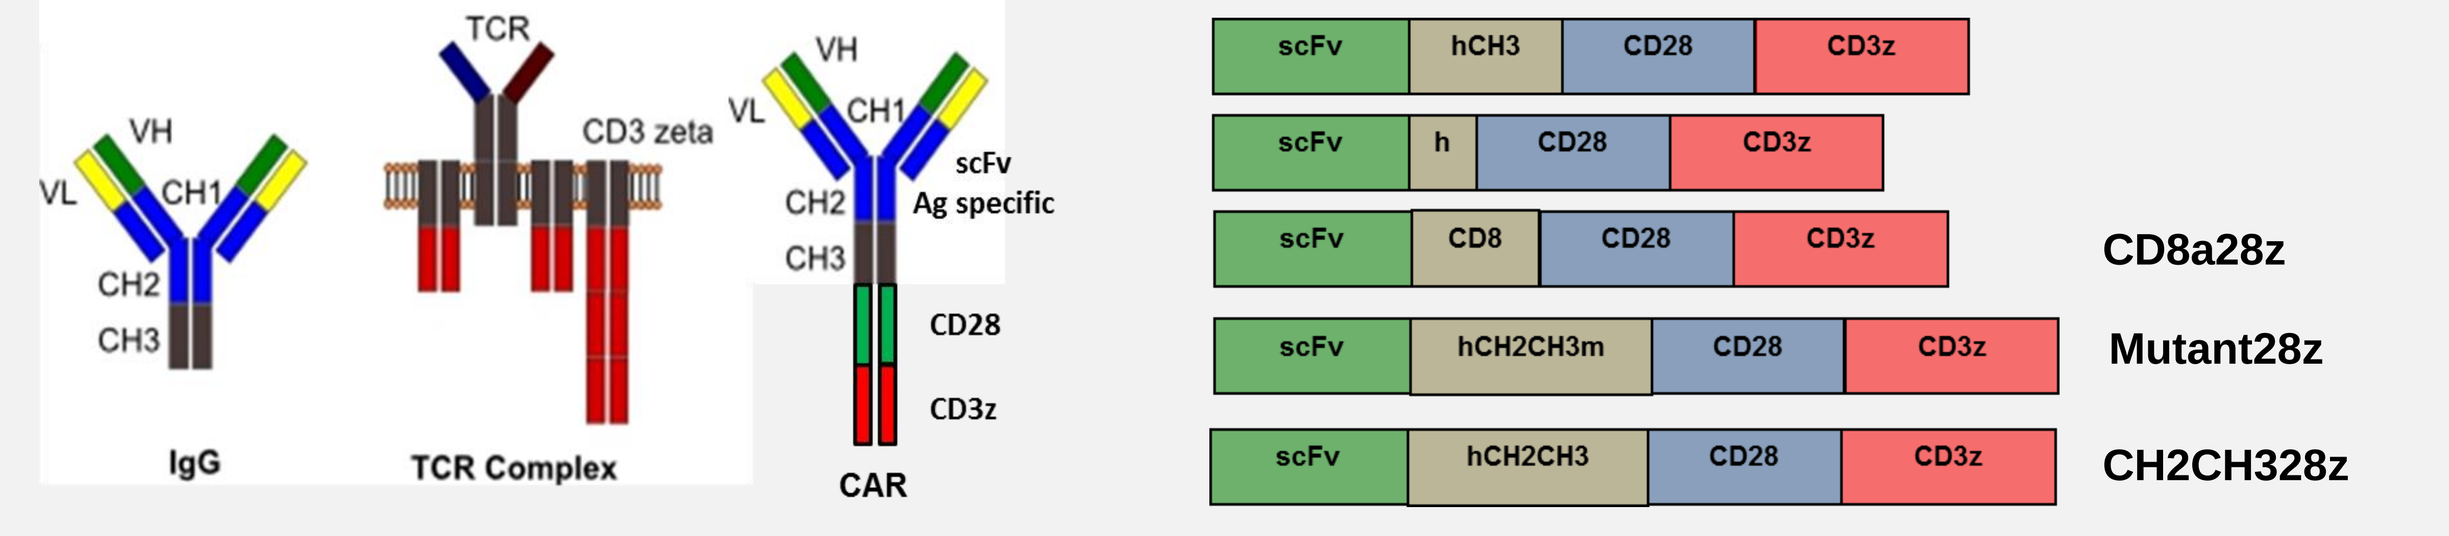
IgG
TCR Complex
CAR
scFv hCH3 CD28 CD3z
scFv h CD28 CD3z
scFv CD8 CD28 CD3z CD8a28z
scFv hCH2CH3m CD28 CD3z Mutant28z
scFv hCH2CH3 CD28 CD3z CH2CH328z

Second generation KMA CAR constructs used in the study with the various spacer regions- CH2CH3 (from IgG1) or mutant CH2CH3 (mutated at the Fcγ binding region) or from CD8α. Six constructs were analysed in detail in this study.

scFv from Kappa Mab (MDX-1097) was fused to CD28 or 41BB co-stimulatory domain and activating ζ subunit of CD3 to generate KMA CAR

RESULTS – KMA CAR T-cells expand in response to KMA

(A) KMA CAR expansion

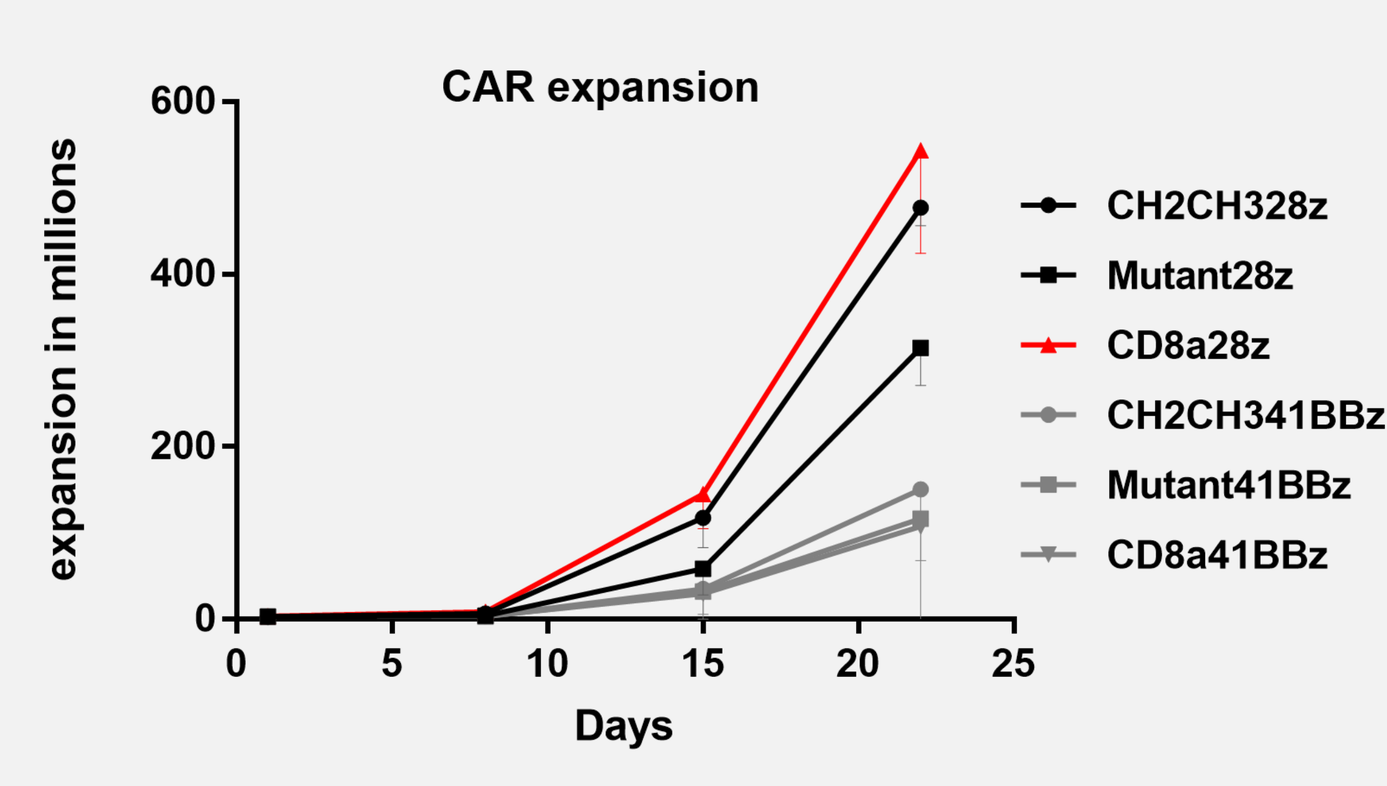
CAR expansion
expansion in millions
Days
CH2CH328z
Mutant28z
CD8a28z
CH2CH341BBz
Mutant41BBz
CD8a41BBz

(B) KMA CAR expression

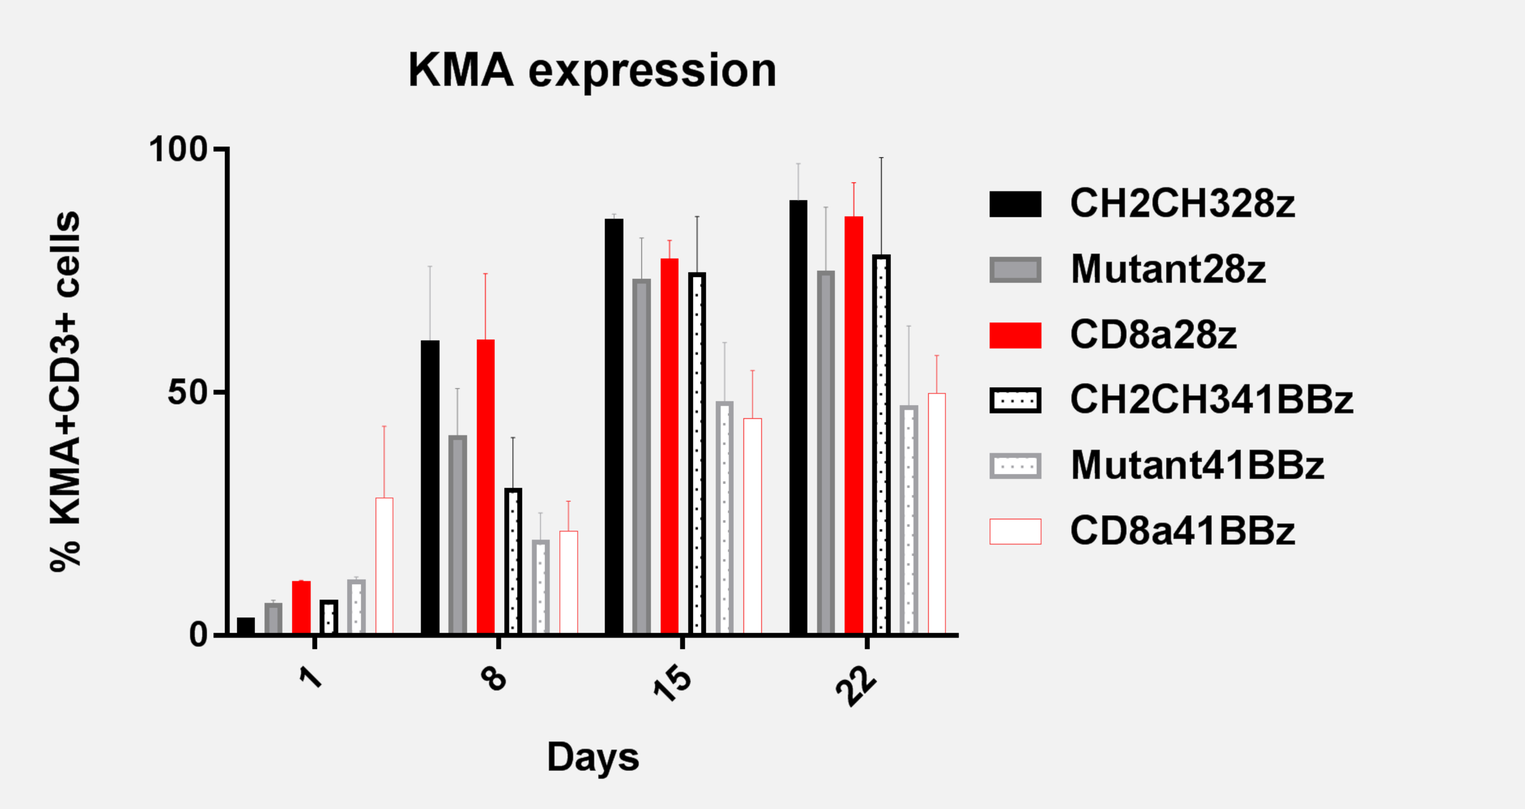
KMA expression
% KMA+CD3+ cells
Days
CH2CH328z
Mutant28z
CD8a28z
CH2CH341BBz
Mutant41BBz
CD8a41BBz

(C) KMA CAR detection

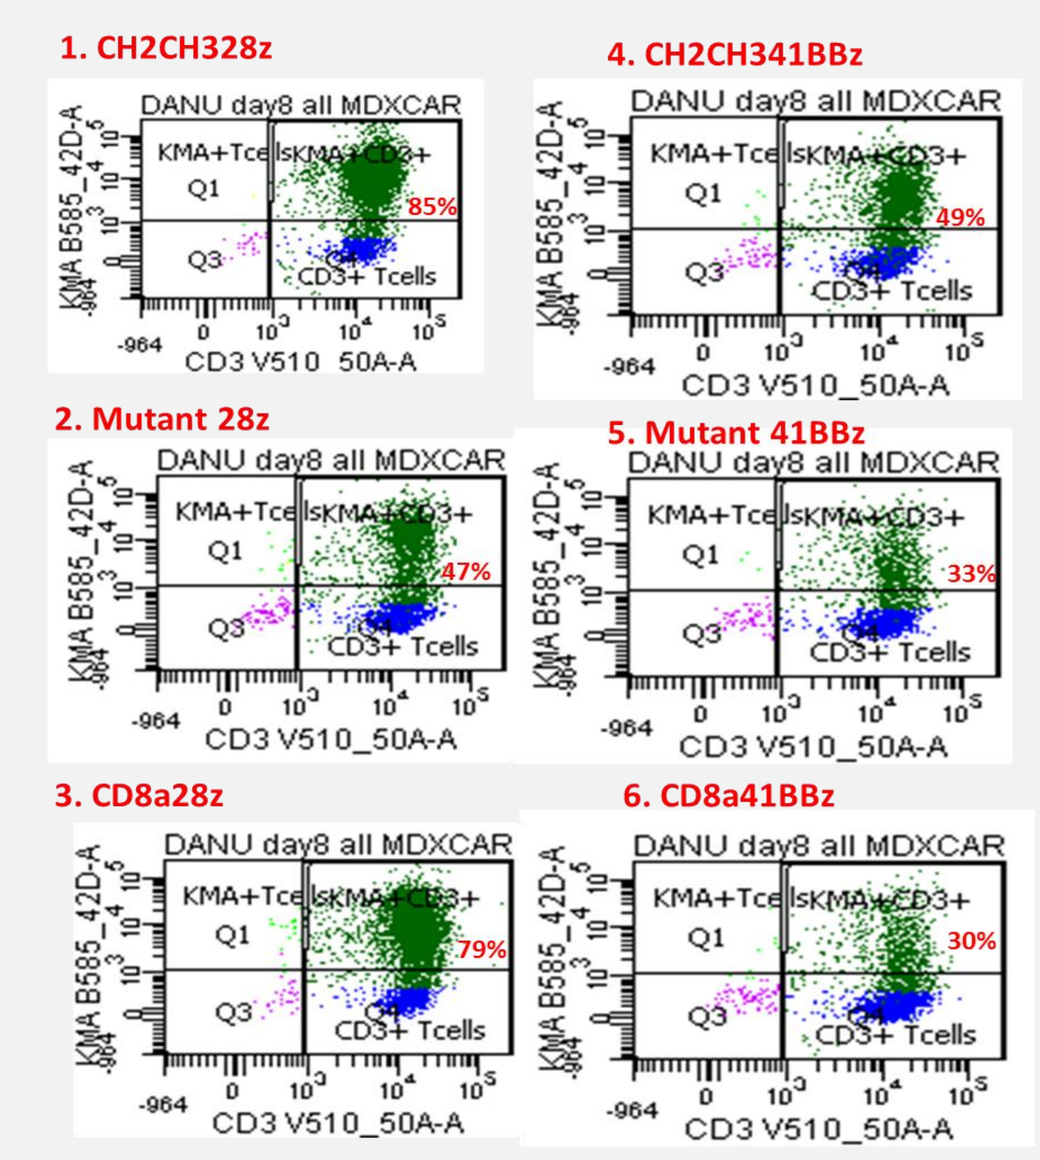
1. CH2CH328z
2. Mutant 28z
3. CD8a28z
4. CH2CH341BBz
5. Mutant 41BBz
6. CD8a41BBz
DANU day8 all MDXCAR
85%
47%
79%
49%
33%
30%

(D) CD8/CD4 distribution

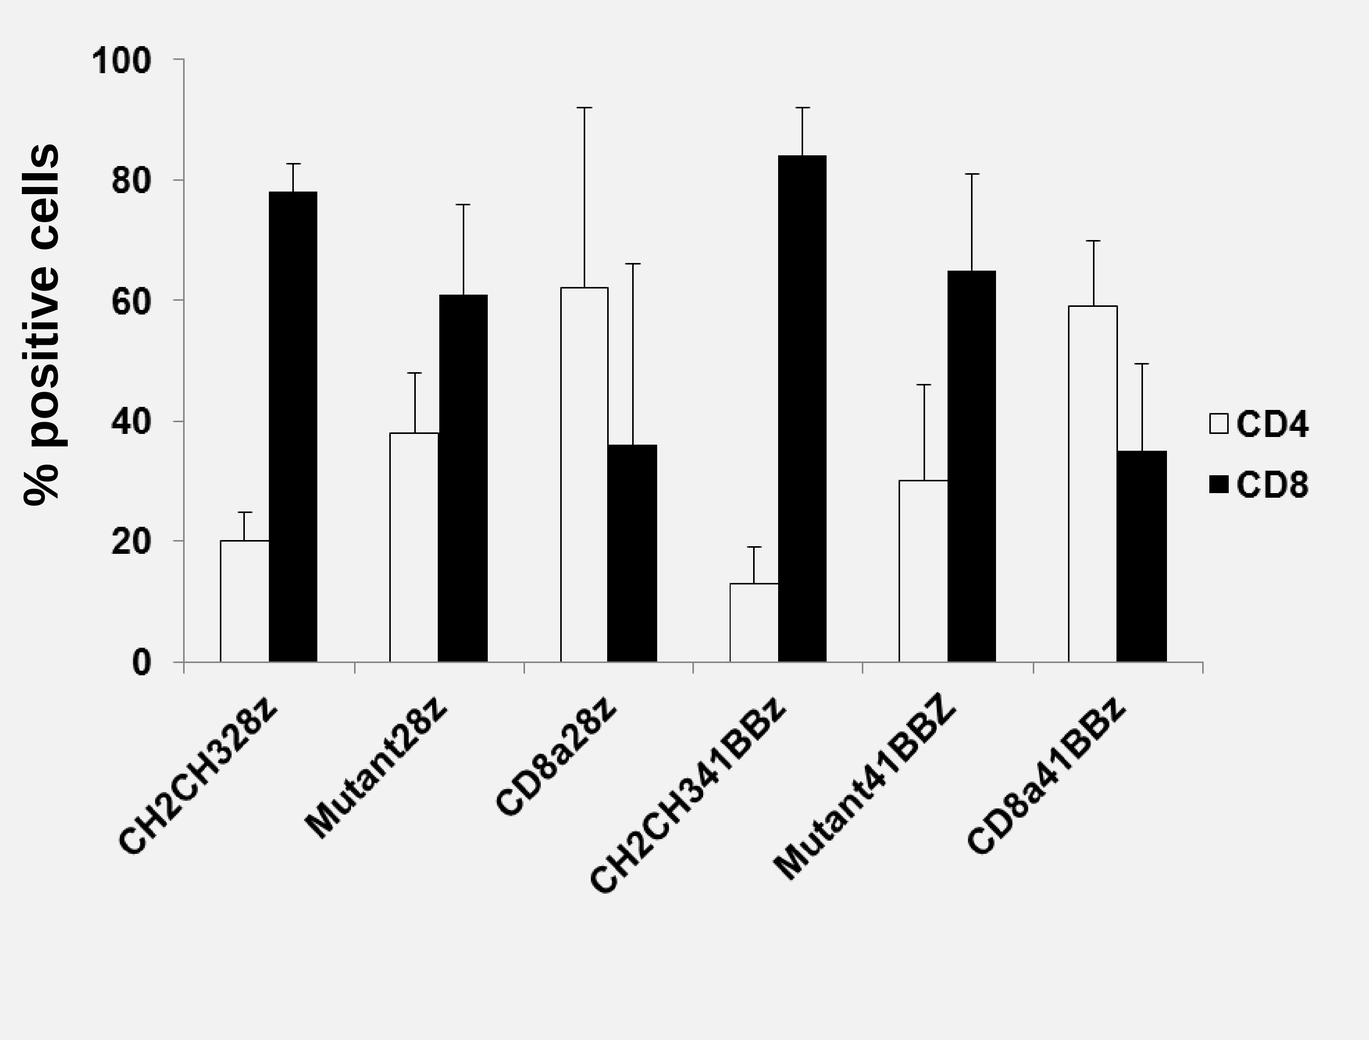
% positive cells
CD4
CD8
CH2CH328z
Mutant28z
CD8a28z
CH2CH341BBz
Mutant41BBZ
CD8a41BBz

***Fig1**(A)*: Average expansion of CAR T-cells from 3 donor PBMCs over 3 weeks showed a range between 150 to 500 fold expansion upon exposure to KMA antigen (CD8α spacer containing CARs in red) *(B&C):* Expression of CAR was detected by staining with biotinylated KMA followed by Streptavidin-PE. % of CAR+ T-cells is plotted (n=3). Flow cytometry dot plots from a representative day 8 culture are shown *(D)* Average CD4/CD8 distribution within the final CAR T-cell culture showing a preponderance of CD8+ T-cells over CD4+ cells in a majority of cultures (n=3).

(E) Memory phenotype

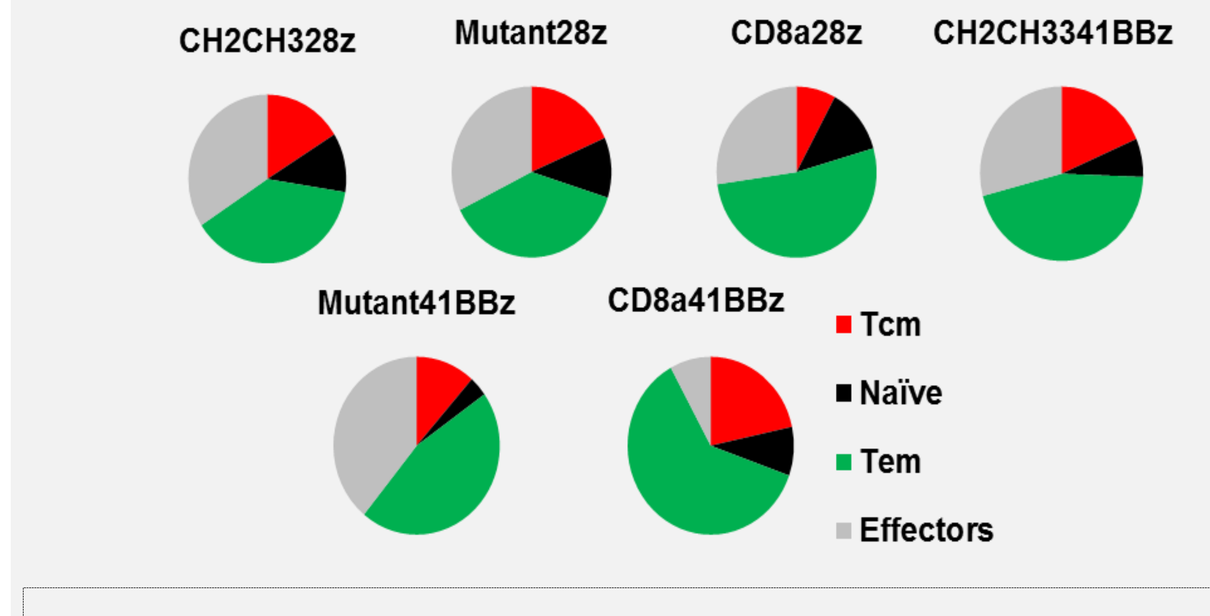
CH2CH328z
Mutant28z
CD8a28z
CH2CH3341BBz
Mutant41BBz
CD8a41BBz
Tcm
Naïve
Tem
Effectors

(F) Exhaustion markers

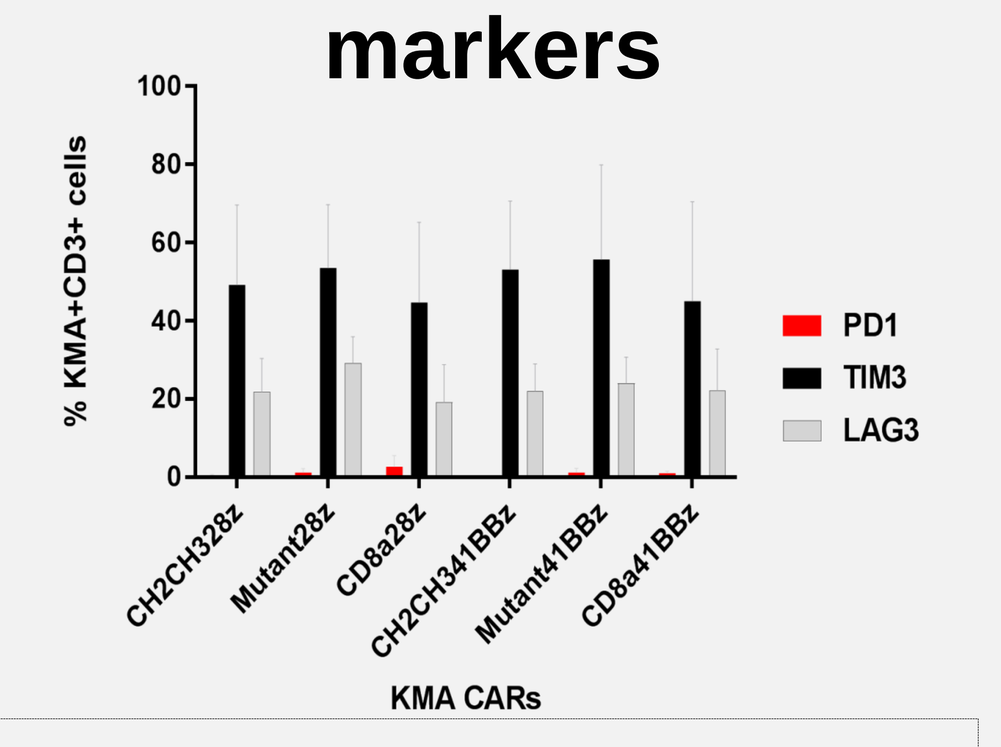
% KMA+CD3+ cells
PD1
TIM3
LAG3
KMA CARs

(G) Cytokine array

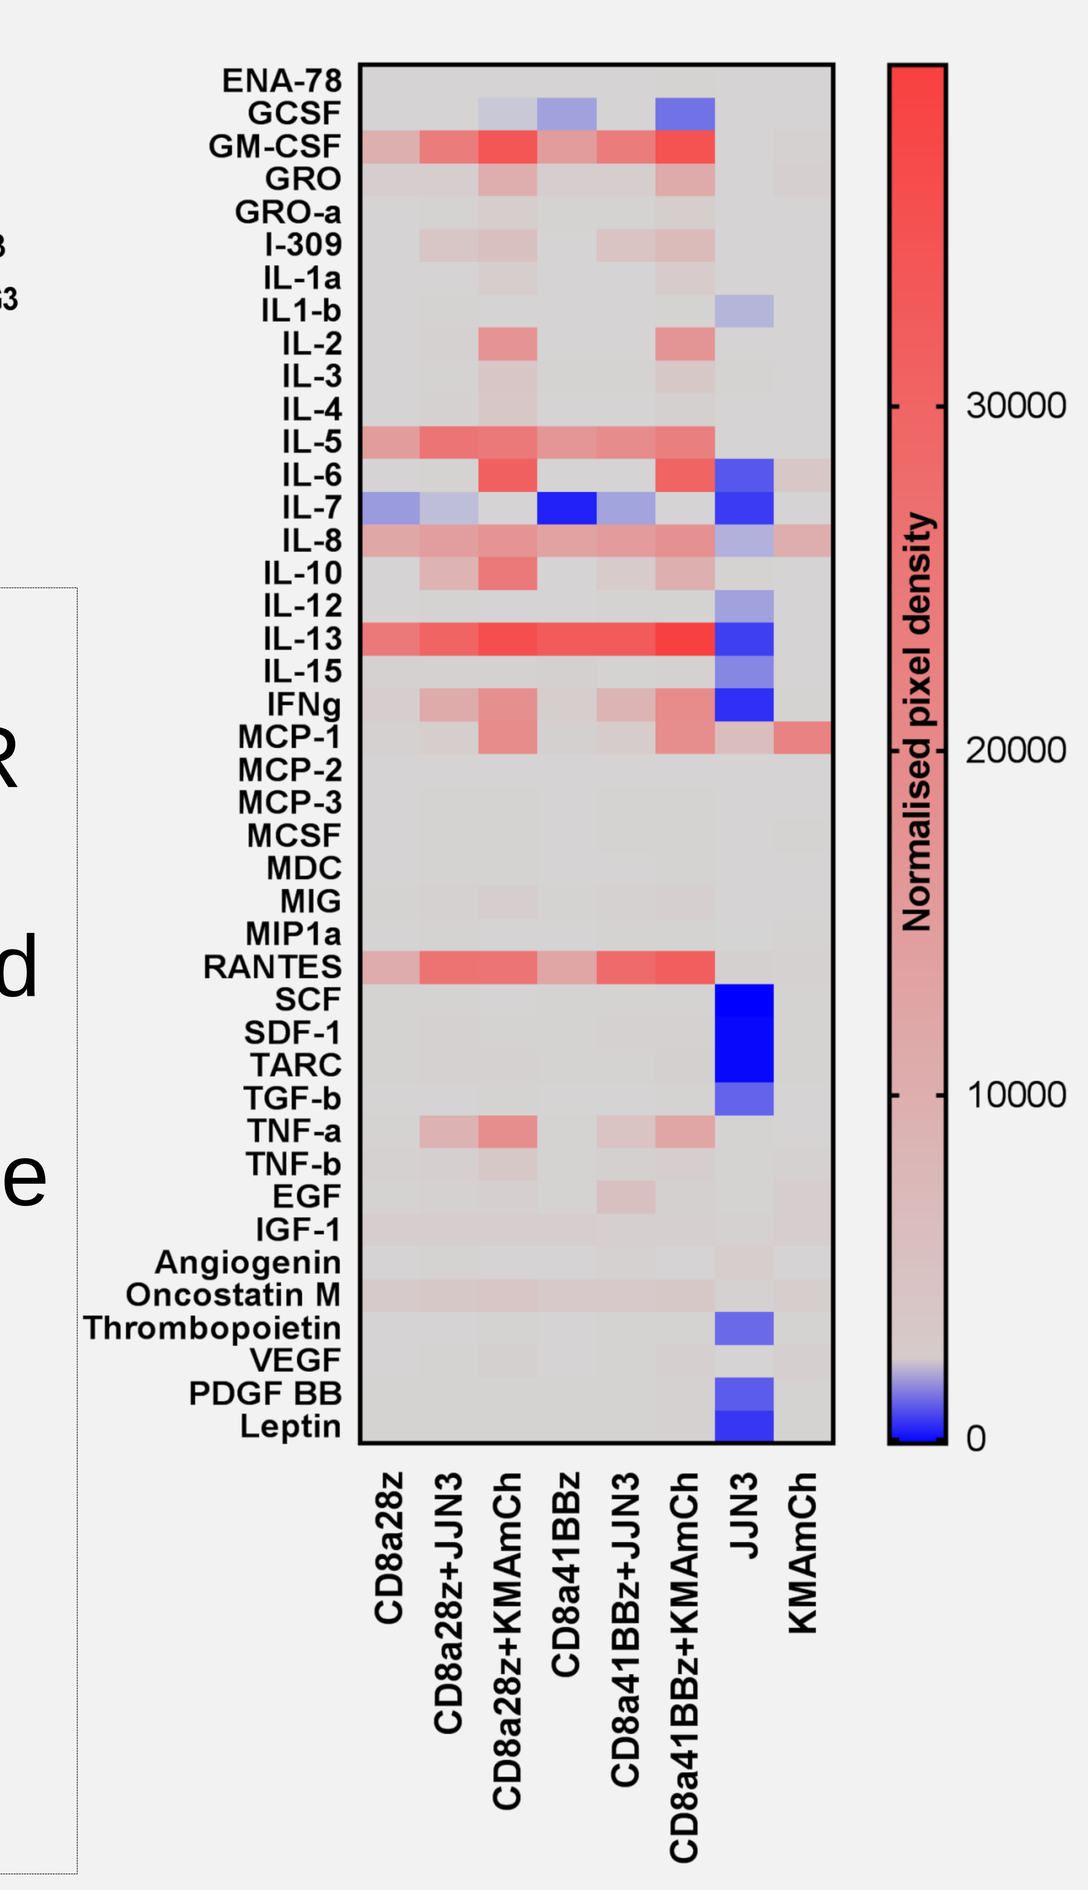
Normalised pixel density
CD8a28z
CD8a28z+JJN3
CD8a28z+KMAmCh
CD8a41BBz
CD8a41BBz+JJN3
CD8a41BBz+KMAmCh
JJN3
KMAmCh

(E) Distribution of naïve, central and effector memory, and 45RA-effector cells is shown after staining of CAR T-cell cultures (d22) with CD45RA and CD62L n=3; *(F)* Expression of exhaustion markers PD-1, TIM-3 and LAG3 on CAR+CD3+ cells was determined by flow cytometry n=3 *(G)* Cytokine membrane array- Release of cytokines from a KMA CD8α28z and CD8α41BBz CAR T-cell culture alone or after co-culture overnight with KMA+cell lines JJN3 and KMA mCherry K562. Cytokines from target cells alone is also indicated. Normalised pixel density representing levels of cytokines is shown in the heat map.

KMA CAR T-cells are specific in their functional response

(A) IFNγ and TNFα release

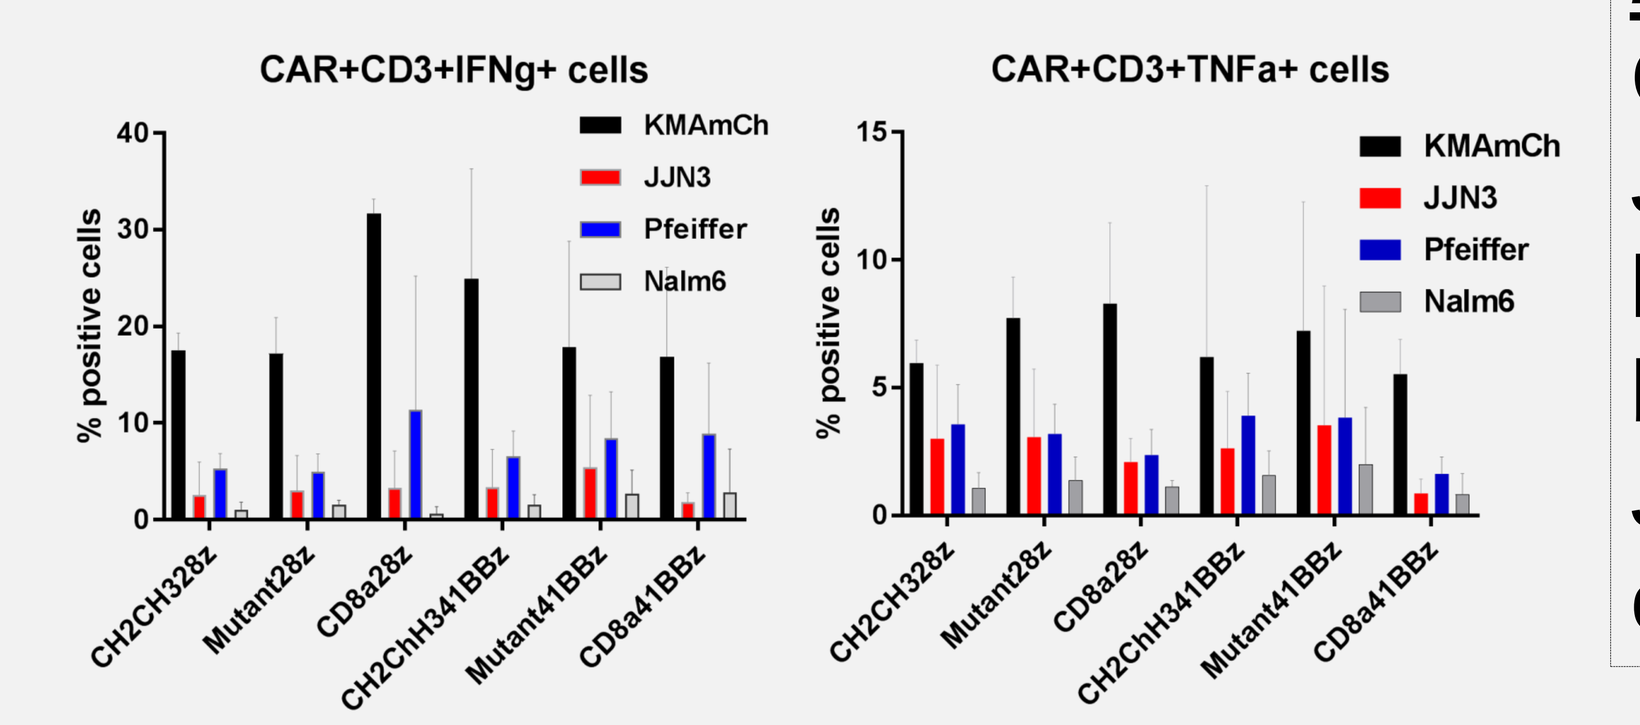
CAR+CD3+IFNg+ cells
CAR+CD3+TNFa+ cells
% positive cells
KMAmCh
JJN3
Pfeiffer
Nalm6

***Fig2** (A)* Average IFNγ and TNFα release from the CAR T-cells only upon co-culture with KMA+ cell lines JJN3 and Pfeiffer and not with KMA-ve CD19+ cell line Nalm6; n=3 *(B)* Cellular cytoxicity as measured by lysis and ^{51}Cr release from KMA+ target cell lines JJN3 and KMA mCherry K562 and not Nalm6 after 4h of co-culture from 3 donor cultures.

(B) ^{51}Cr release measuring cytotoxicity

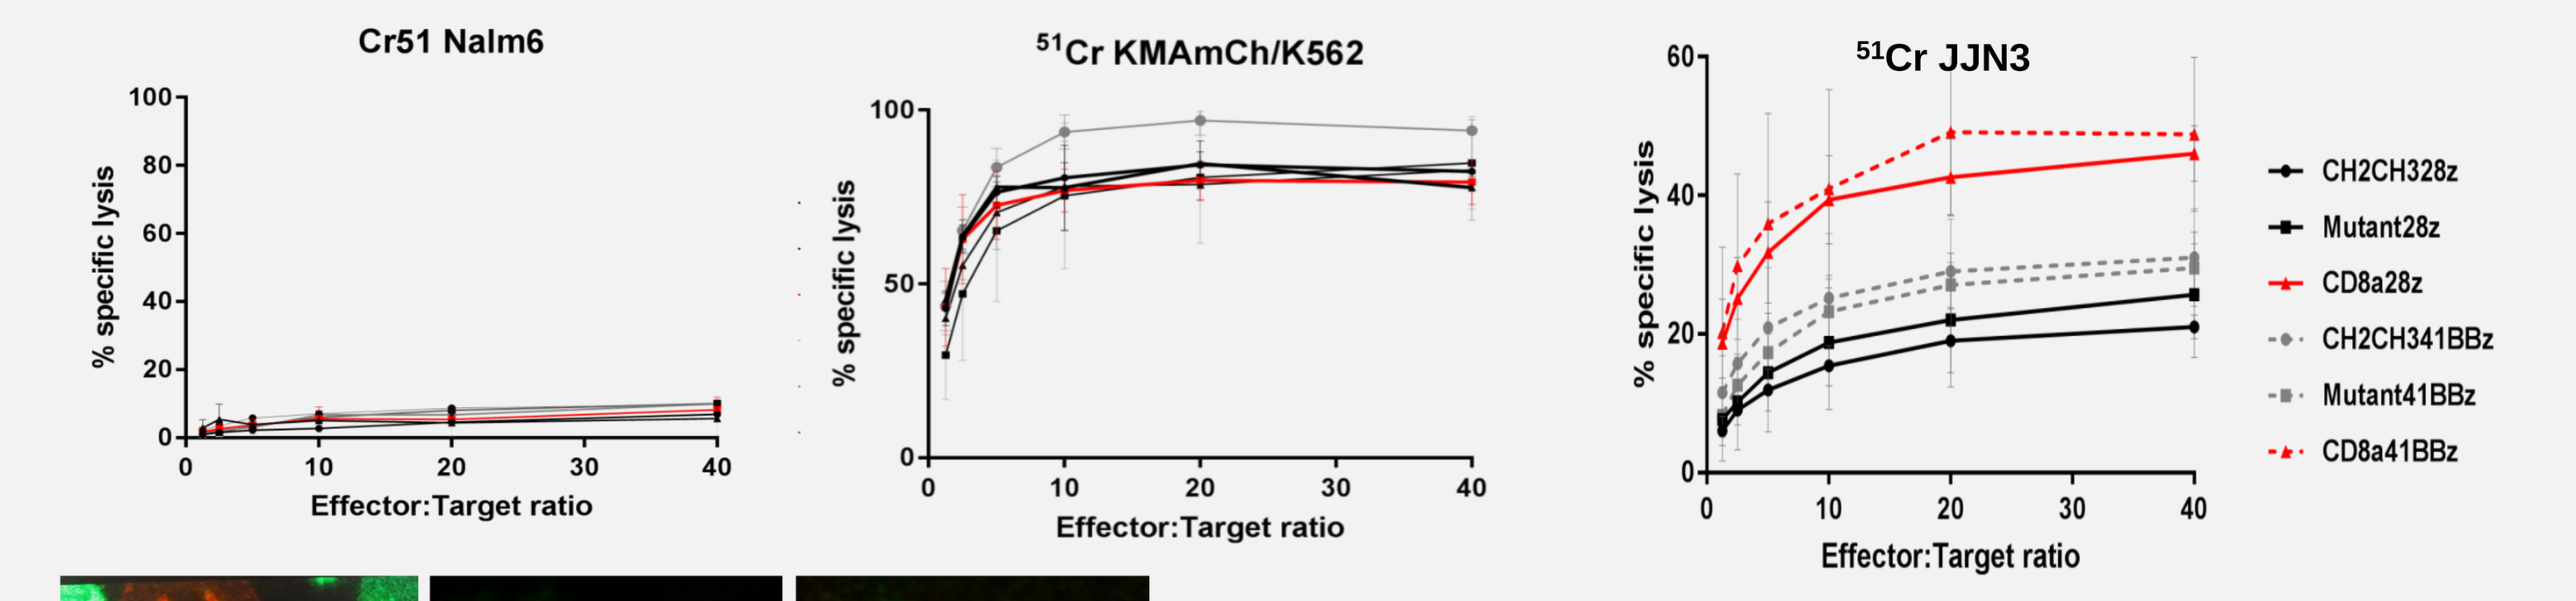
Cr51 Nalm6
51Cr KMAmCh/K562
51Cr JJN3
% specific lysis
Effector:Target ratio

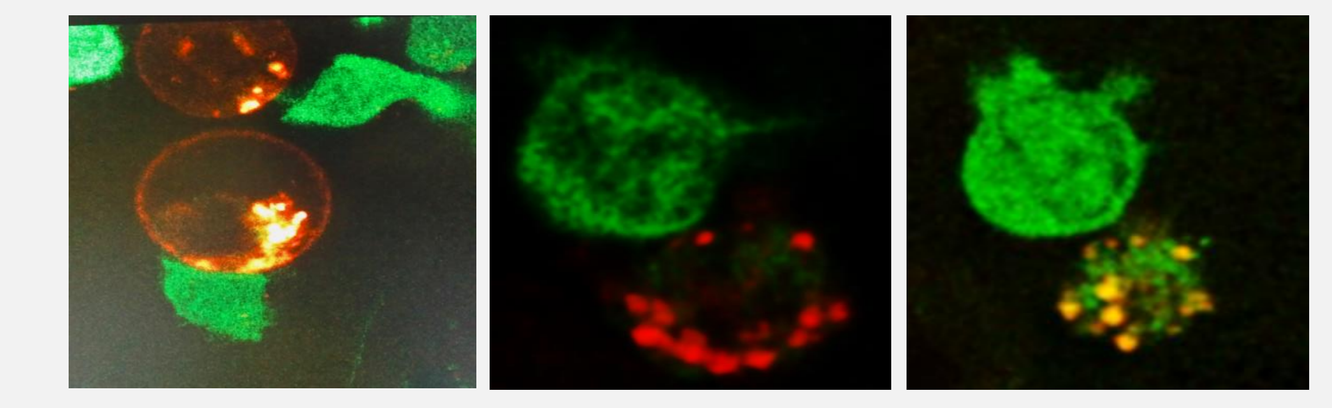

Fig2C: Representative confocal fluorescence images of GFP tagged green CD8α28z CAR T-cells attacking the target KMA mCherry K562 cells after 4h of co-culture.

CAR T-cell function is not inhibited by free κ-light chain

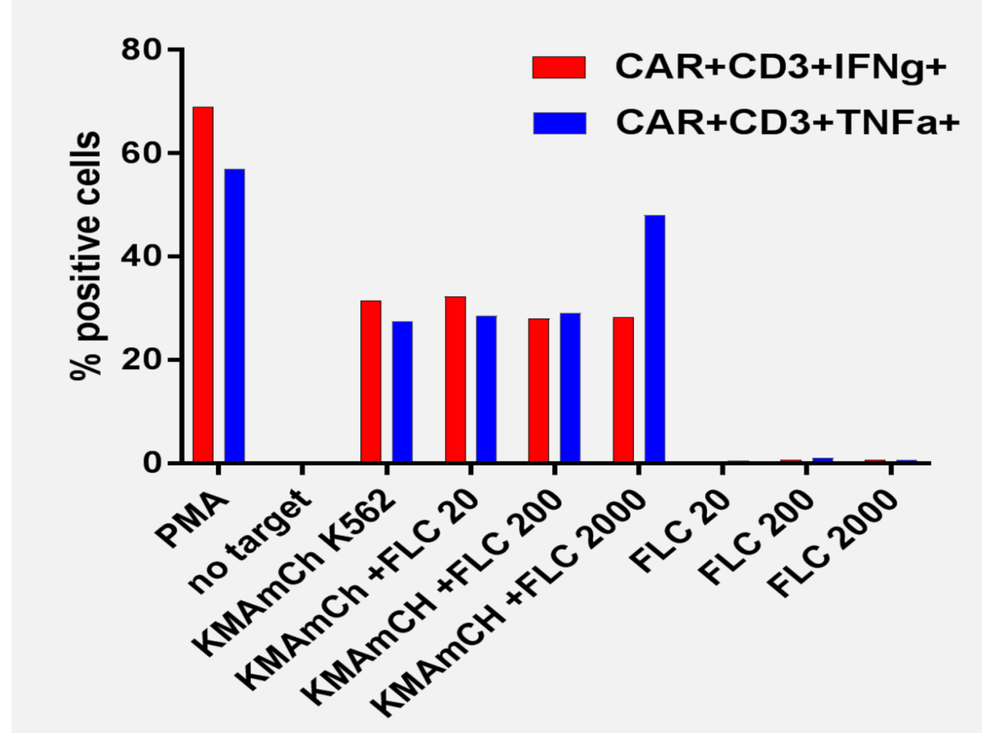
CAR+CD3+IFNg+
CAR+CD3+TNFa+
% positive cells
PMA
no target
KMAmCh K562
KMAmCh +FLC 20
KMAmCH +FLC 200
KMAmCH +FLC 2000
FLC 20
FLC 200
FLC 2000

Fig3: A representative CD8α28z KMA CAR T-cell culture specifically releasing IFNγ and TNFα when co-cultured with KMA mCherry K562. This release was not inhibited when free κ-light chains at 20, 200, 2000mg/l were added to the co-culture. Free κ-light chains by themselves (FLC 20,200, 2000 lanes) did not elicit a response similar to the negative control when no target cells were added. PMA served as a positive control for the cytokine release assay.

SUMMARY

- **CAR T-cells targeting KMA are highly specific and effective**
- **KMA CAR T-cells containing the CD8α spacer expanded the best *in vitro* and were the most efficacious against KMA+ cells**
- 28z co-stimulatory domain containing CAR T-cells expanded better *in vitro* than the 41BB co-stimulatory domain containing CAR T-cells
- Approximately 30% of the CAR+T-cells were memory T-cells with negligent expression of PD-1 and expected to persist and function *in vivo*; However, expression of TIM-3 and LAG-3 was significant
- CAR T-cell function is not inhibited by the presence of free kappa light chains
- ***In vivo* testing in Rag2-/- mouse models and primary tumour cells currently underway**

SIGNIFICANCE

- Myeloma is the next haematological malignancy after leukaemia that is showing phenomenal response to CAR T-cell therapy.
- KMA is a highly specific antigen present on malignant cells and not on normal plasma cells or normal haematopoeitic stem cells, making it an ideal targetable antigen.